ECCMID 2017
Poster P0469
22 April 2017

Efficacy of ASN100, a combination of two human monoclonal antibodies neutralizing six *Staphylococcus aureus* cytotoxins, in a CA-MRSA rabbit necrotizing pneumonia model

Lukas Stulik[1], Delphine Labrousse[2], Delphine Croisier[2], Gábor Nagy[1], Eszter Nagy[1]
[1] Arsanis Biosciences GmbH, Vienna, Austria, [2] Vivexia Laboratory, Dijon, France

Introduction

Cytotoxins of *Staphylococcus aureus* are implicated in the pathogenesis of pneumonia. Necrotizing pneumonia, a particularly severe form associated with high mortality, is most commonly caused by community-associated methicillin-resistant *S. aureus* (CA-MRSA). The role of alpha-haemolysin (Hla) is well established in *S. aureus* pneumonia pathogenesis, and the contribution of bi-component leukocidins (such as the Panton-Valentine Leukocidin [PVL]) has been shown in rabbits that are, unlike mice, sensitive to all five bi-component leukocidins produced by *S. aureus*. ASN100 is an equimolar combination of two human monoclonal antibodies (ASN-1 and ASN-2) that together neutralize six cytolysins (ASN-1: Hla, HlgAB, HlgCB, LukED and LukSF; ASN-2: different LukGH/LukAB variants). ASN100 was tested for efficacy in a lethal USA300 CA-MRSA rabbit necrotizing pneumonia model. ASN100 is currently being studied in a Phase 2 clinical trial for the prevention of pneumonia in mechanically-ventilated patients who are heavily colonized with *S. aureus*.

Methods

Male New Zealand White rabbits were prophylactically treated with ASN100 at doses ranging from 0.08 to 20 mg/kg (six rabbits in each of the five dose groups) 24 hours prior to intratracheal instillation of a lethal dose of a PVL+ USA300 CA-MRSA strain (LAC). Survival was monitored for up to 156 hours post challenge. Gross necropsy, lung histopathology, and bacterial organ load analyses were performed in a blinded fashion at 12 hours post challenge. Statistical analyses: Kaplan-Meier plots with log-rank (Mantel-Cox) test; Group comparison by Mann-Whitney test.

Dose-dependent protection by ASN100

24 hours prior to lethal, intratracheal challenge with the USA300 CA-MRSA strain, rabbits were prophylactically treated with different doses of ASN100 and survival was subsequently monitored up to 156 hours post challenge.

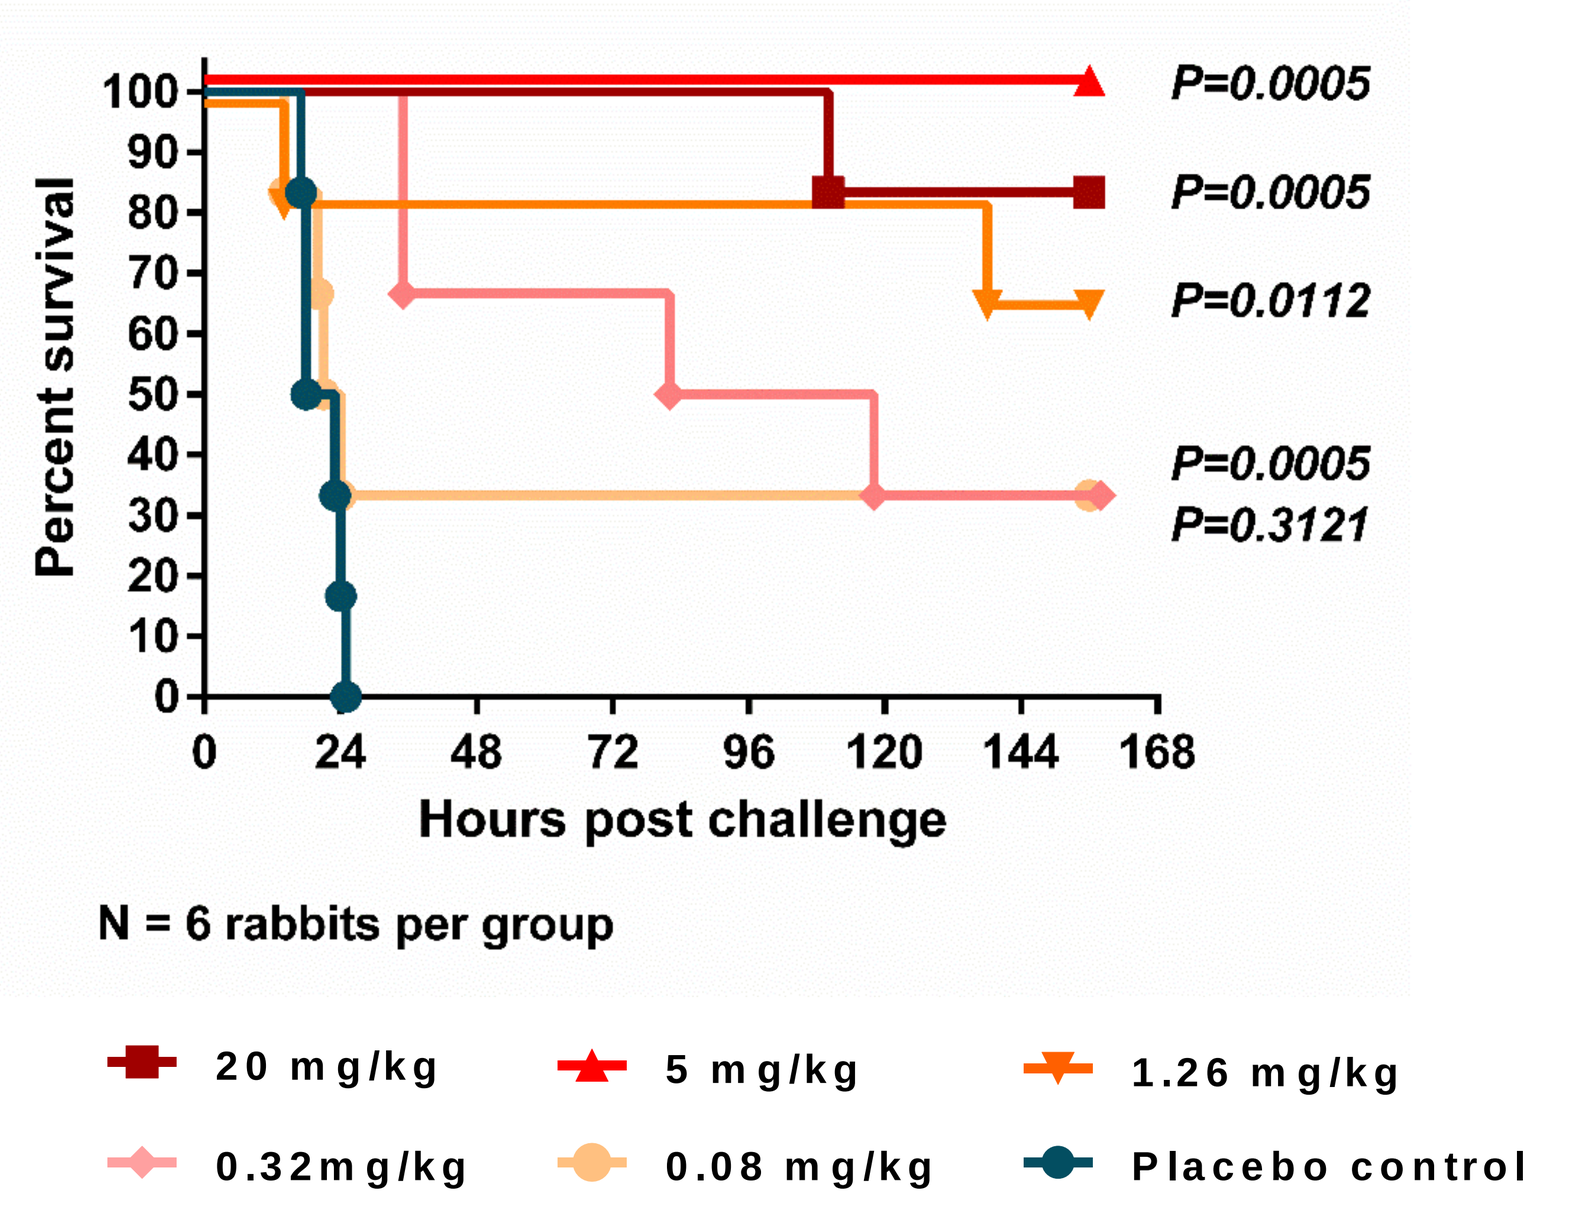
P=0.0005
P=0.0005
P=0.0112
P=0.0005
P=0.3121
Percent survival
Hours post challenge
N = 6 rabbits per group
20 mg/kg
5 mg/kg
1.26 mg/kg
0.32mg/kg
0.08 mg/kg
Placebo control

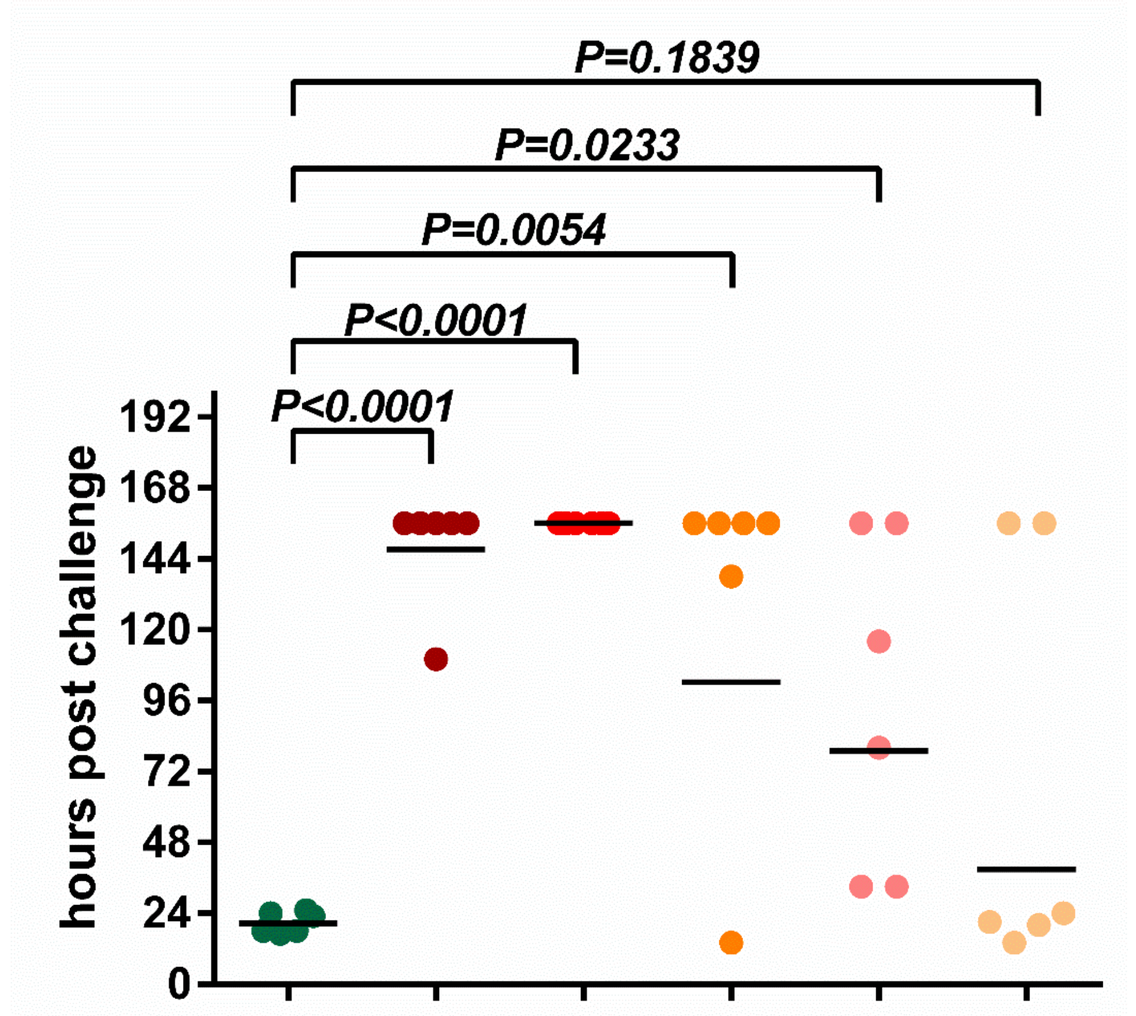
P=0.1839
P=0.0233
P=0.0054
P<0.0001
P<0.0001
hours post challenge

ASN100 protected rabbits from lethal necrotizing pneumonia in a dose-dependent manner.

Doses as low as 0.32 mg/kg significantly improved survival of animals challenged with the USA300 CA-MRSA, which is also reflected by the time-to-death analysis.

ASN100 reduces the bacterial burden in different organs

The three highest doses of ASN100 (1.26-20 mg/kg) were evaluated for their contribution to improve the bacterial burden at early stages of infection. Rabbits were sacrificed at 12 hours post intratracheal challenge with the USA300 CA-MRSA.

Lung

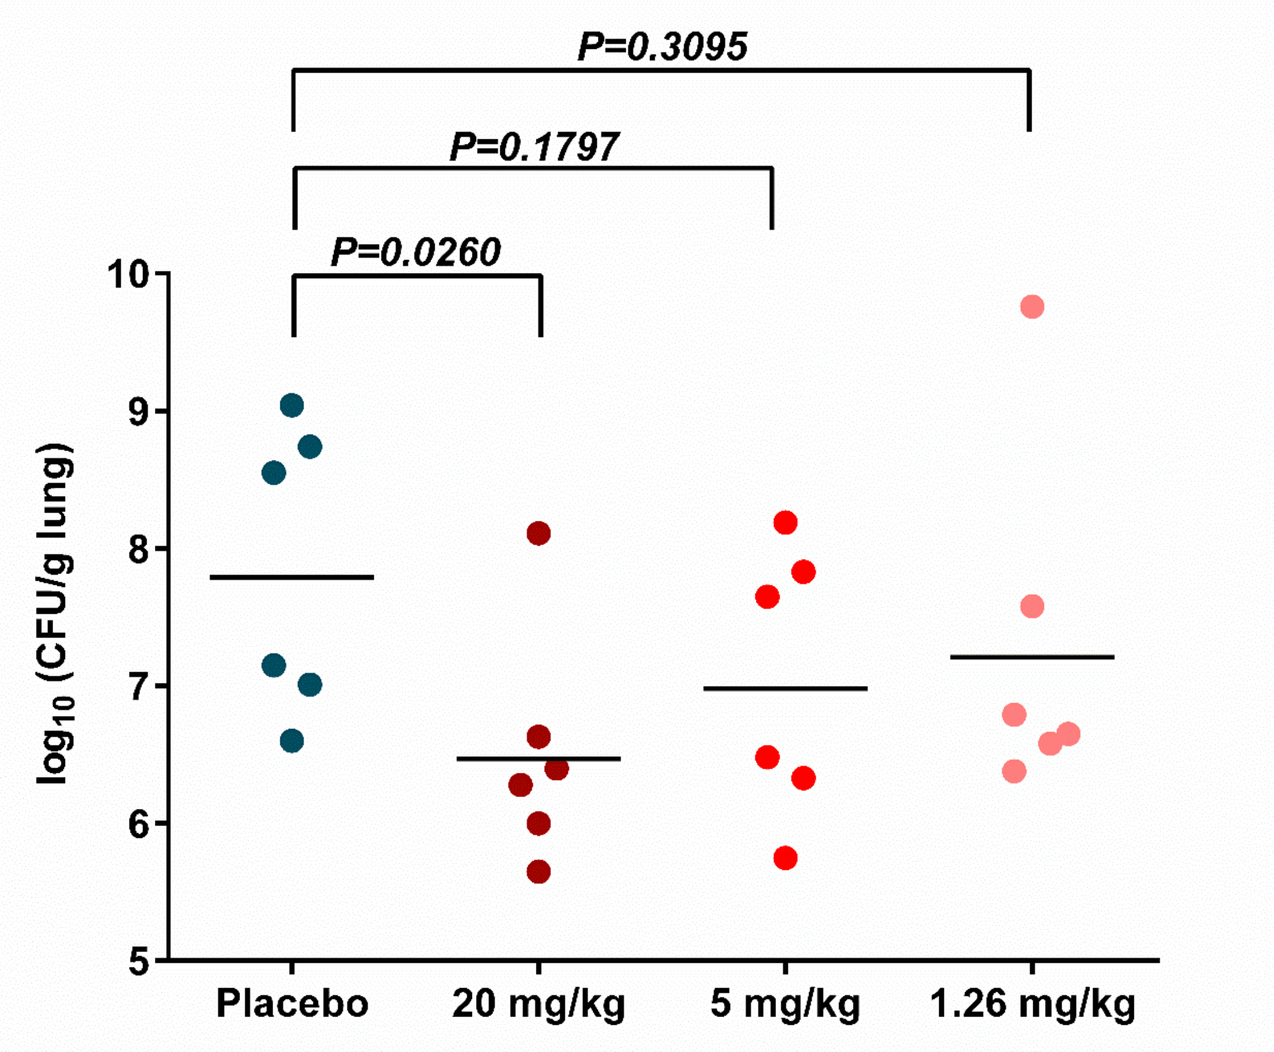
P=0.3095
P=0.1797
P=0.0260
log10 (CFU/g lung)
Placebo
20 mg/kg
5 mg/kg
1.26 mg/kg

Spleen

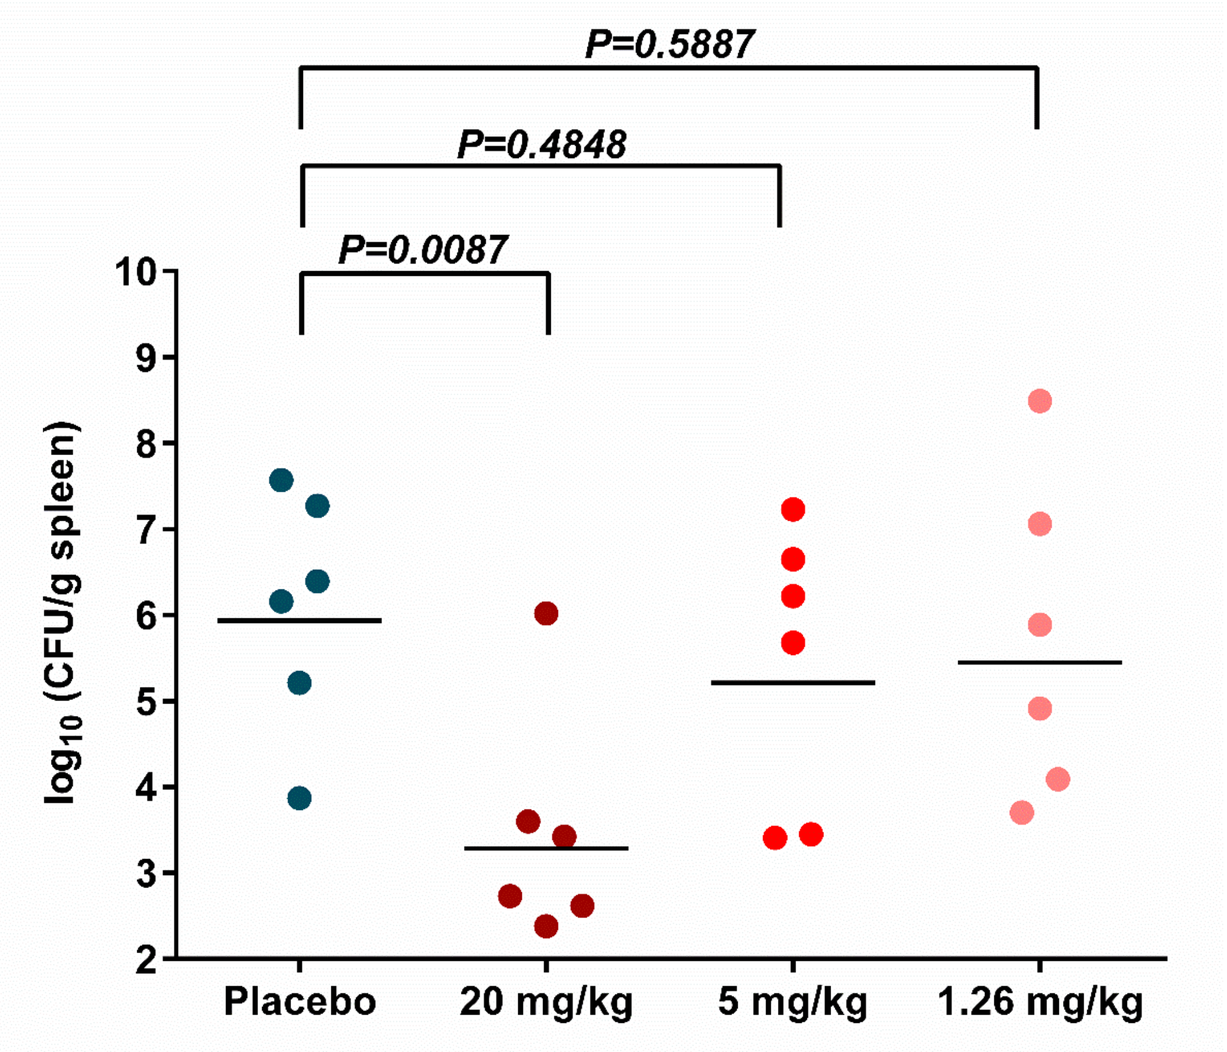
P=0.5887
P=0.4848
P=0.0087
log10 (CFU/g spleen)
Placebo
20 mg/kg
5 mg/kg
1.26 mg/kg

Kidneys

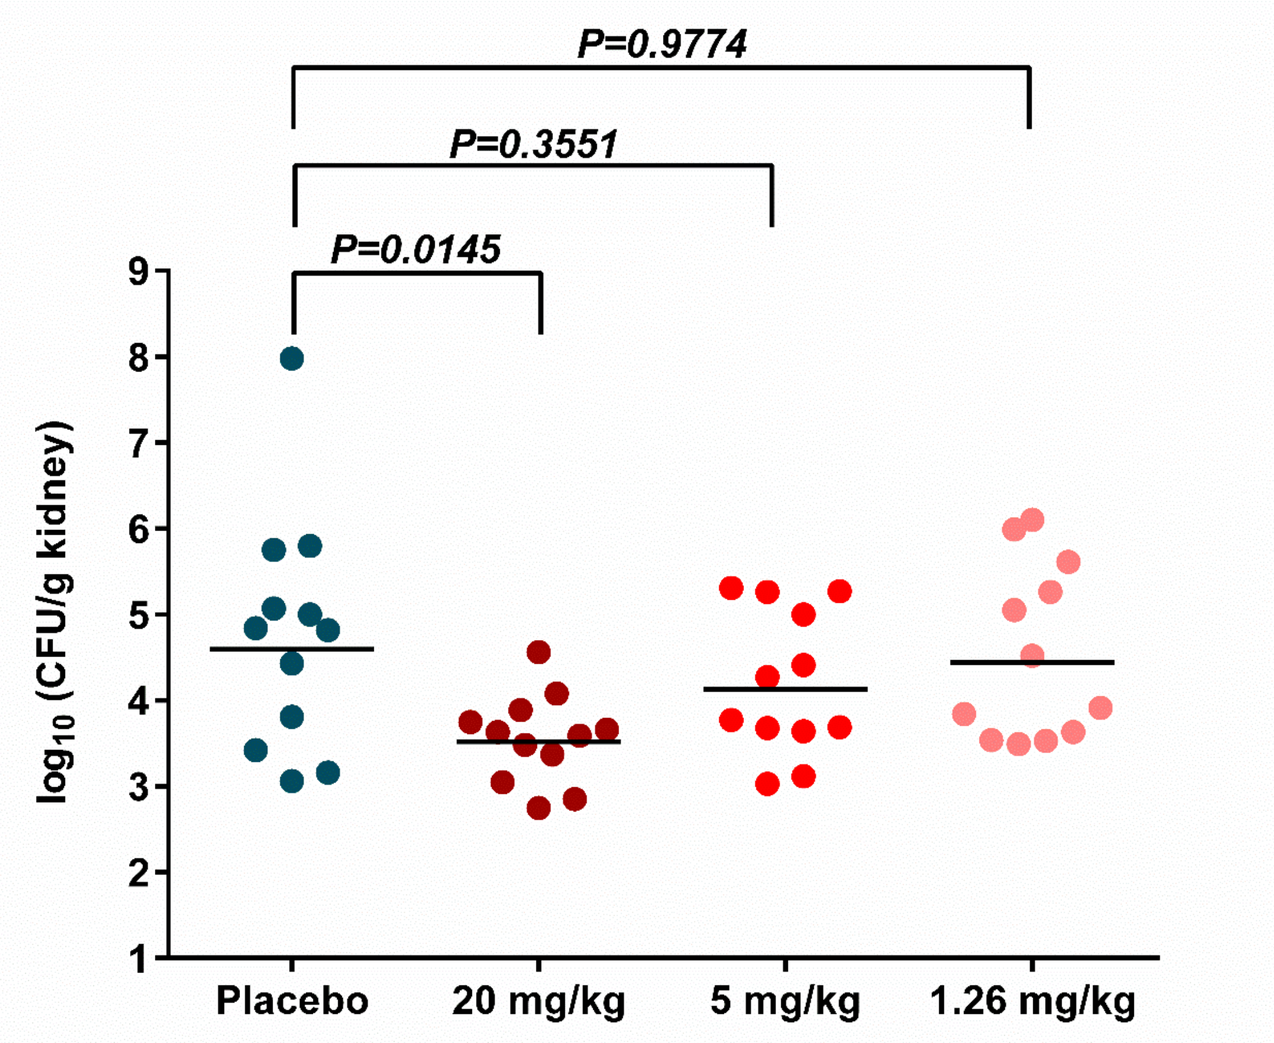
P=0.9774
P=0.3551
P=0.0145
log10 (CFU/g kidney)
Placebo
20 mg/kg
5 mg/kg
1.26 mg/kg

ASN100 significantly reduced the bacterial burden in the lungs and systemically at the highest mAb dose, already at early stages of infection.

Toxin neutralization improves macroscopic lung pathology

The lung pathology of infected animals was evaluated at 12 hours post challenge by means of macroscopic gross-necropsy.

Placebo

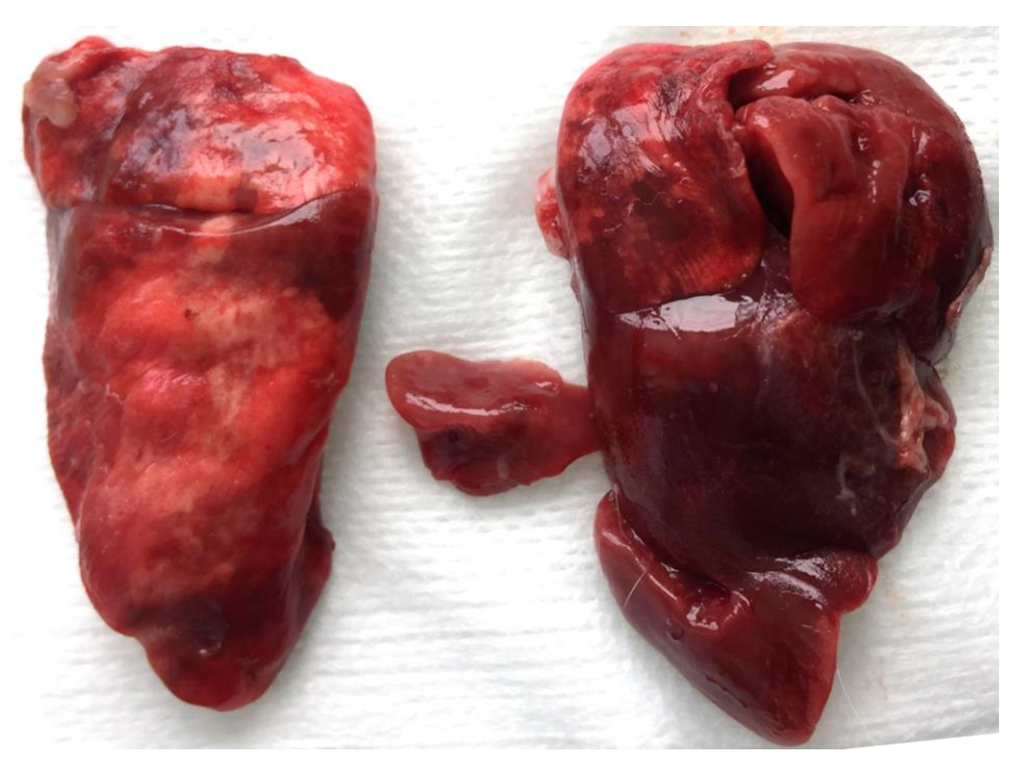

ASN100 (20 mg/kg)

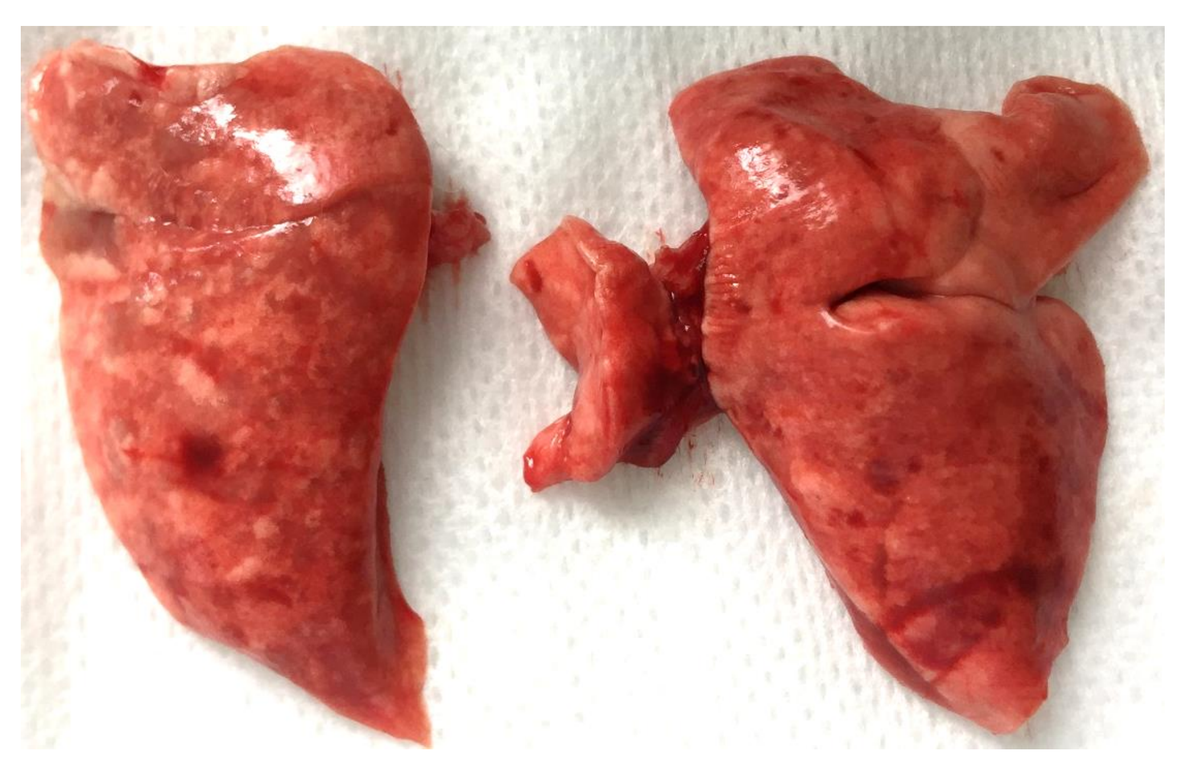

Macroscopic lung score

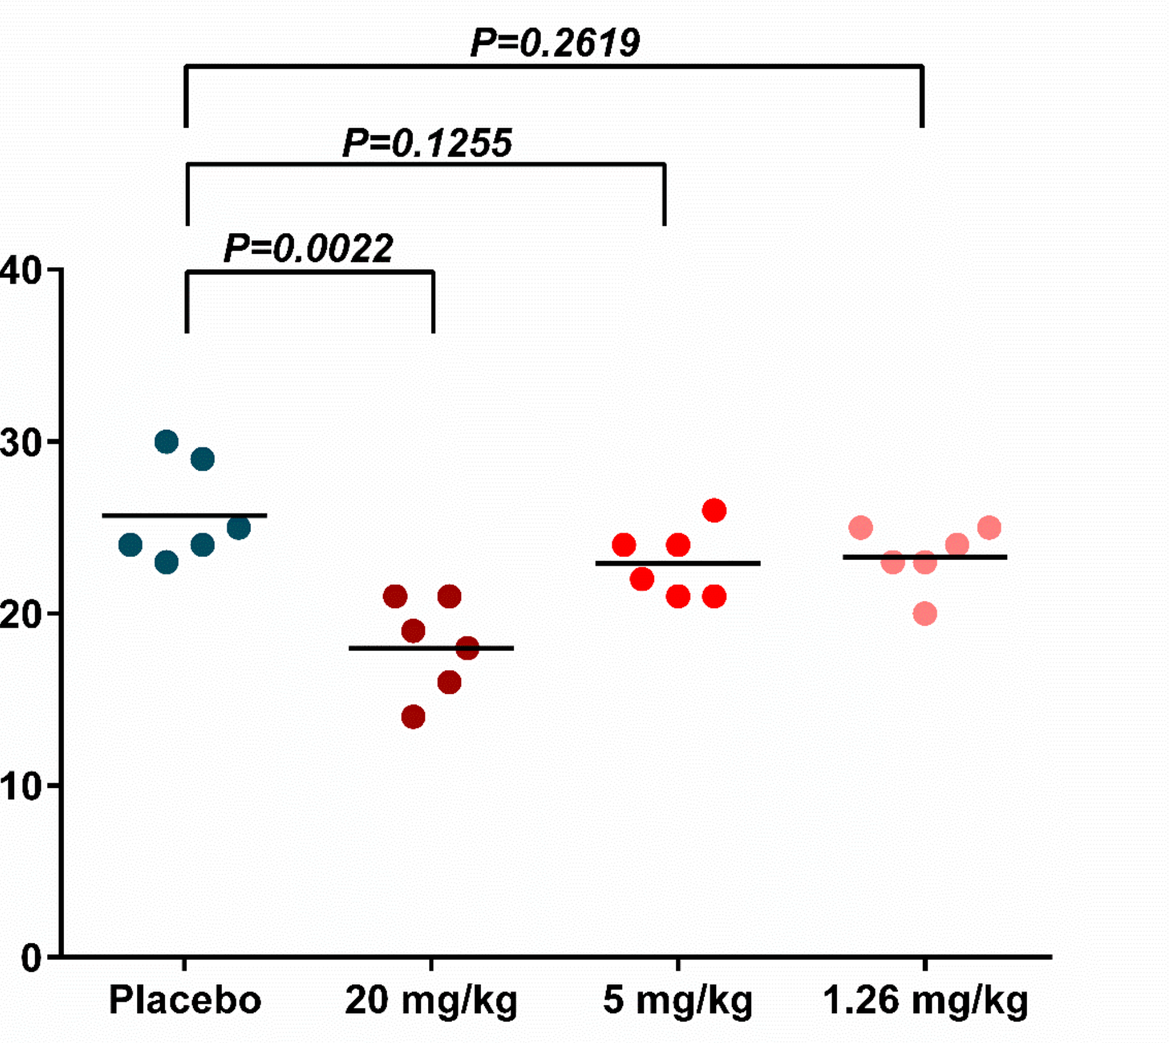
P=0.2619
P=0.1255
P=0.0022
Macroscopic lung score
Placebo
20 mg/kg
5 mg/kg
1.26 mg/kg

Lung oedema rate

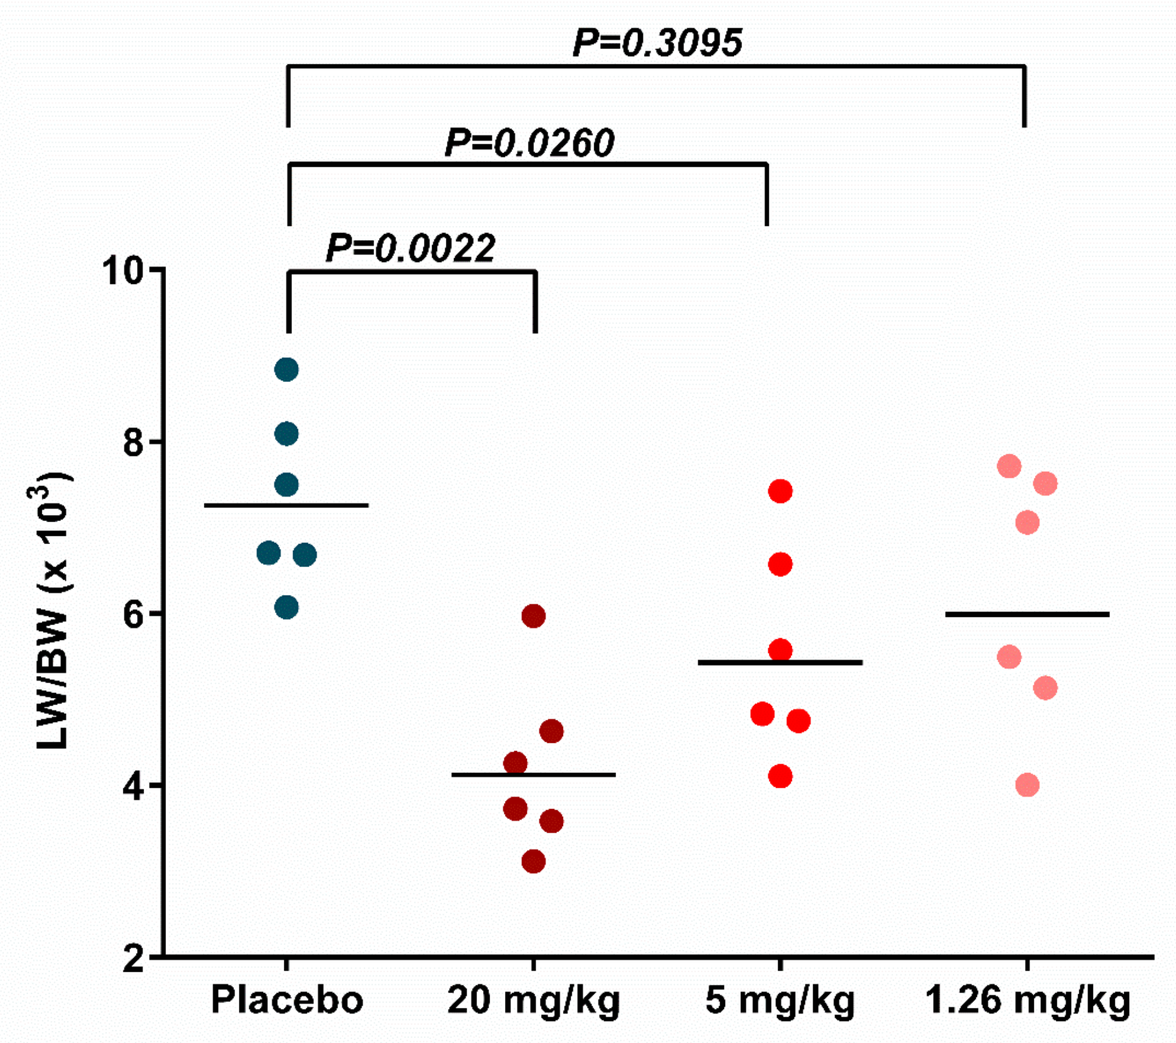
P=0.3095
P=0.0260
P=0.0022
LW/BW (x 10³)
Placebo
20 mg/kg
5 mg/kg
1.26 mg/kg

Lung tissue damage was prevented by ASN100 and also the lung oedema rate improved, reaching significance at the highest dose of ASN100.

ASN100 preserves the lung architecture and immune cells

The lung pathology of infected animals was evaluated at 12 hours post challenge by histopathology (H&E and Giemsa staining of central lung lobes).

Placebo | ASN100 (20 mg/kg)

H&E

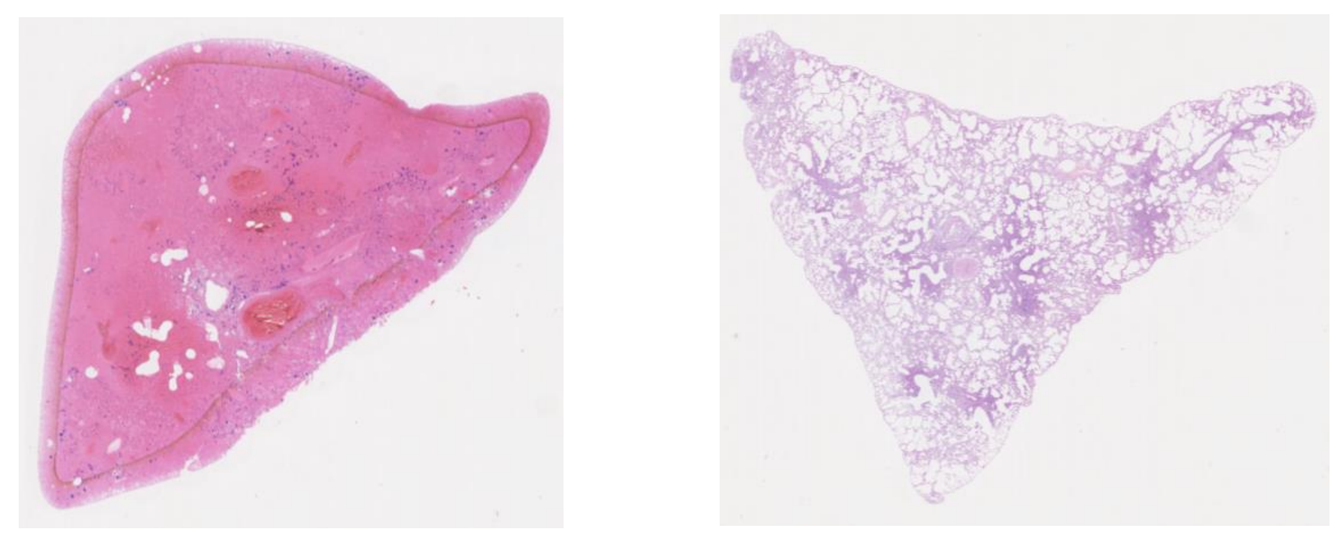

Giemsa

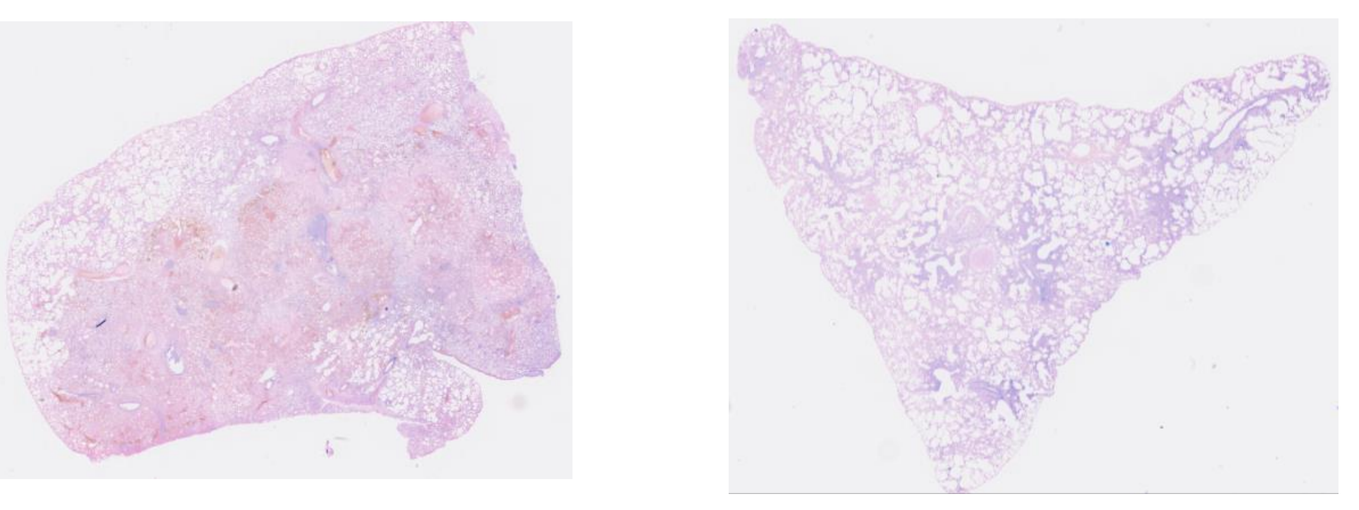

Lung score without immune cells

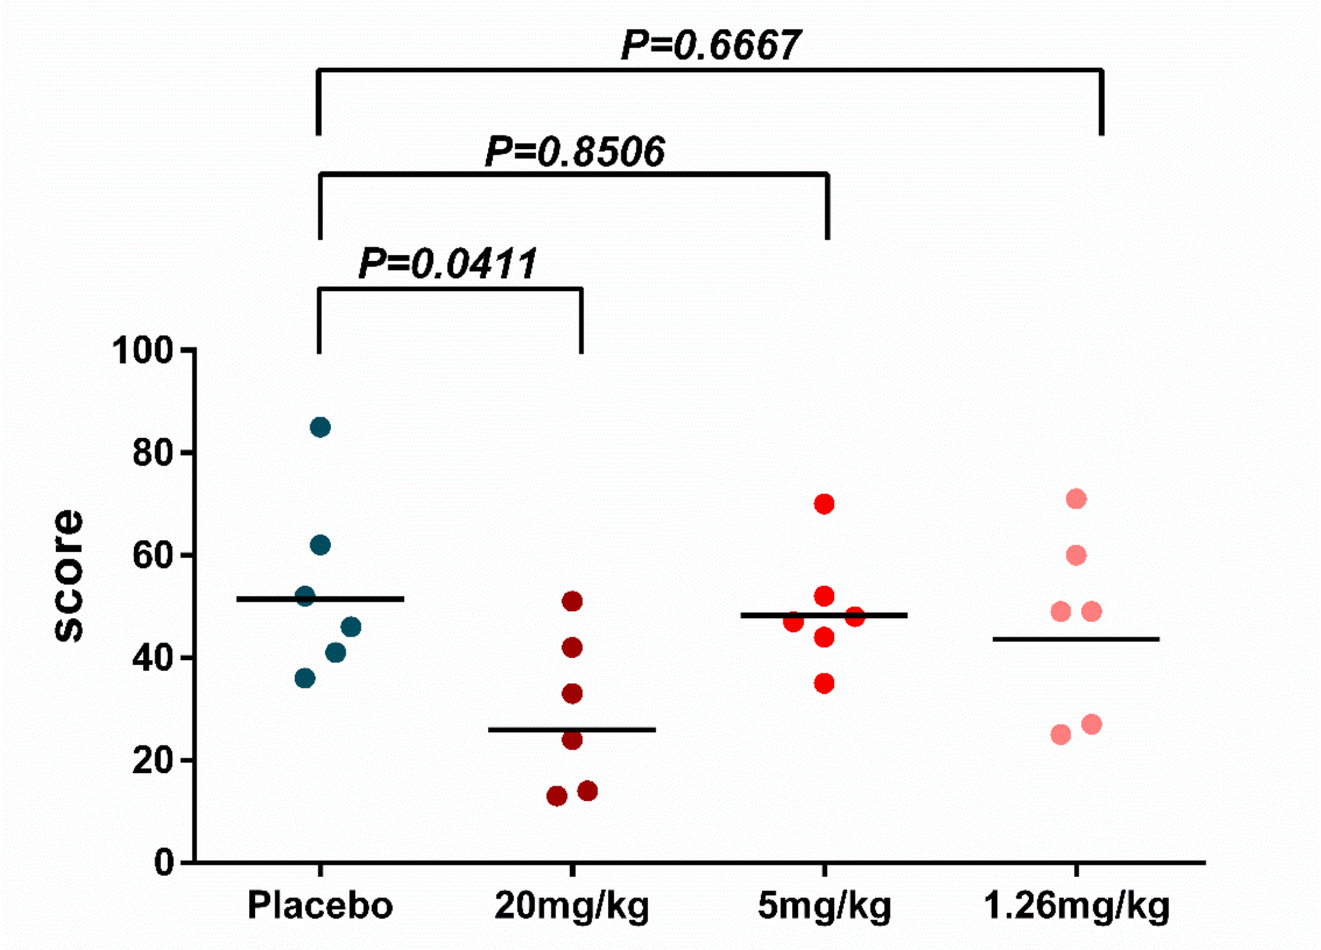
P=0.6667
P=0.8506
P=0.0411
score
Placebo
20mg/kg
5mg/kg
1.26mg/kg

Lung macrophages and neutrophils

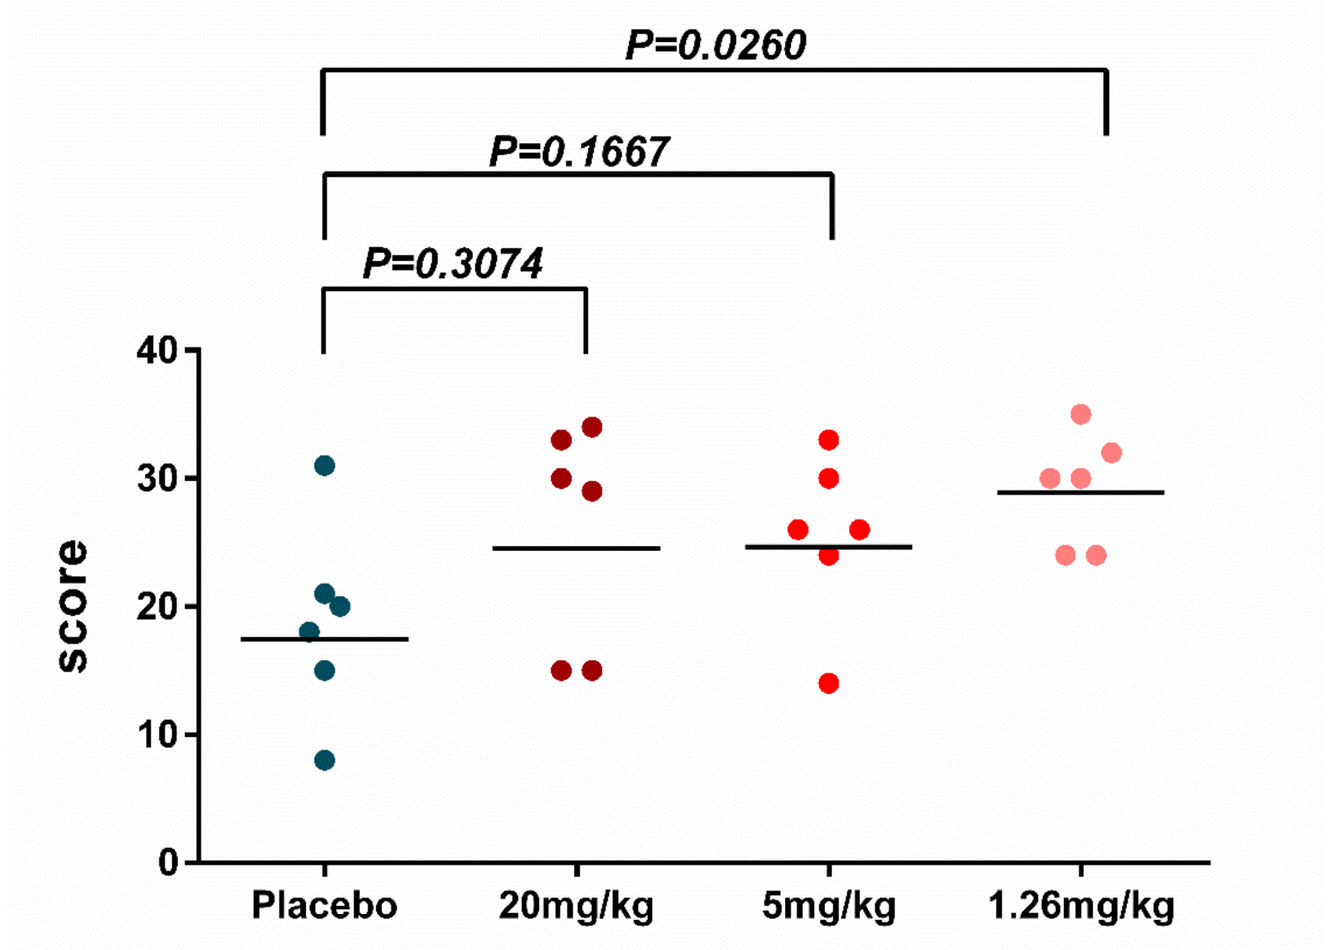
P=0.0260
P=0.1667
P=0.3074
score
Placebo
20mg/kg
5mg/kg
1.26mg/kg

The histopathological lung score correlated well with the macroscopic score and revealed a preservation of macrophages and neutrophils, showing this tendency at all tested dose-levels of ASN100.

Conclusions and Outlook

- ASN100, a combination of two human monoclonal antibodies neutralizing six *S. aureus* cytolysins, elicited a significant protective efficacy in this lethal rabbit model of necrotizing pneumonia caused by a USA300 CA-MRSA.
- The observed dose-dependent survival benefit was associated with a reduced bacterial burden in lungs and distal organs as well as a signifcantly improved lung pathology at the highest dose tested at early stages of infection.
- Ongoing preclinical studies are designed to corroborate this potent efficacy of ASN100 against a range of additional *S. aureus* isolates in such models.
- ASN100 is currently tested in a Phase 2 clinical trial, evaluating the prevention of *S. aureus* pneumonia in heavily colonized, mechanically ventilated patients.

Acknowledgements

We thank Sandrine Albac and Davy Hayez for their technical support.

Disclosure and Contact

Disclosure: All authors are employees and shareholders in the companies performing this work.

Contact: Lukas Stulik, Arsanis Biosciences GmbH, Helmut-Qualtinger-Gasse 2, 1030 Vienna, Austria. Email: lukas.stulik@arsanis.com